In vivo delivery of a novel CD3-targeted Lentiviral Vector generates CD19 CAR-T cells in two different humanized mouse models and results in complete B cell depletion

Frederic Vigant[1], Anirban Kundu[1], Dongming Zhang[1], Wei Zhang[1], Ewa Jaruga-Killeen[1], Michelle Andraza[1], Gregory Schreiber[1], Alissa R Kerner[1], Junyi Zhang[1], John Henkelman[1], Renata Soares[1], Ramya Yarlagadda[1], Gregory I Frost[1] & Sid P Kerkar[1]

[1]Exuma Biotech Corporation, West Palm Beach, FL 33401

EXUMA Biotech

3294

Key Messages

1. *In vivo* CD3-targeted Lentivectors (LV) delivery of CAR-T transgenes represents a promising CAR-T gene therapy approach (GCAR)
2. CD3-directed CAR LV generate CD19 CAR-T cells *in vivo,* resulting in a complete depletion of hCD20+ B cells in circulation and in lymphoid organs
3. This novel approach for the rapid and direct formation of CAR-T cells *in vivo* was demonstrated in two different humanized mouse models
4. B-Cell aplasia was obtained in lymphoreplete animal models without the use of a conditioning chemotherapy regimen, consistent with our results from studies of *ex vivo* transduced PBMC with CD3-directed LV

Abstract

Background: Chimeric antigen receptor therapies (CAR-T) are highly effective against hematologic malignancies but require a lymphodepleting chemotherapy regimen and are faced with many challenges including manufacturing time, scalability, and cost of production due to the need for *ex vivo* culturing of cells and complex chain of custody requirements. *In vivo* delivery of self-inactivating lentiviral vectors (LV) encoding for CAR-T transgenes represents a promising strategy to improve the time to treatment, scalability, and cost of current CAR-T therapies.

Methods: Lentiviral vectors encoding a CD19 CAR with a synthetic driver element were manufactured in a chemically defined cell substrate that incorporates a modified envelope designed to target and activate CD3+ T Cells. Two different humanized mouse models were utilized. In a PBMC humanized mouse model, NSG MHC Class I/II knock-out mice (DKO) were injected with 1E7 human PBMC and followed one day later with 2E7 TU of LV encoding for CD19 CAR. In parallel, CD34+ humanized NSG SGM3 mice were also administered 2E7 TU LV encoding CD19 CAR. Flow cytometry of peripheral blood samples was evaluated at various time points for the presence of CAR+ cells and CD20+ B cells. Additionally, tissue samples were examined by histopathology, and PCR for vector copy numbers.

Results: The NSG SGM3 humanized CD34+ mouse model exhibited efficient chimerism of human CD45+ hematopoietic cells (>50% of live cells in peripheral blood). Apart from CD15+ neutrophils, all major human immune cell components were well represented in peripheral blood, including CD14+ Monocytes, CD20+ B cells, and CD3+ Lymphocytes. At study initiation, the T cell compartment in the SGM3 mice exhibited skewing towards CD4+ T cells. Injection of a CD19 CAR-LV resulted in a significant reduction of CD20+ B cells as early as 5 days post-injection. CD3+ CAR+ cells were detected in peripheral blood by day 5 with evidence of CAR+ cell expansion at subsequent time points. Loss of CD20+ B cells was stable throughout the observation period with some mice exhibiting a complete elimination of B cells. Similarly, the PBMC humanized NSG DKO mice injected with CD19 CAR-LV showed evidence of CAR+ cells in peripheral blood.

Conclusion: CD3-directed self-inactivating lentiviral vectors can efficiently deliver an integrating CAR gene into T Lymphocytes following *in vivo* administration. These *in vivo* generated CD19 CAR T cells expand systemically and effectively eliminate pre-existing B cells. These data show that targeting of CD3 through *in vivo* delivery can produce functional CAR T cells and represents an innovative therapeutic opportunity to potentially overcome current manufacturing times, scalability, and cost challenges facing cell therapies.

Results *in vitro*

Figure 1. *In Vitro* Transduction

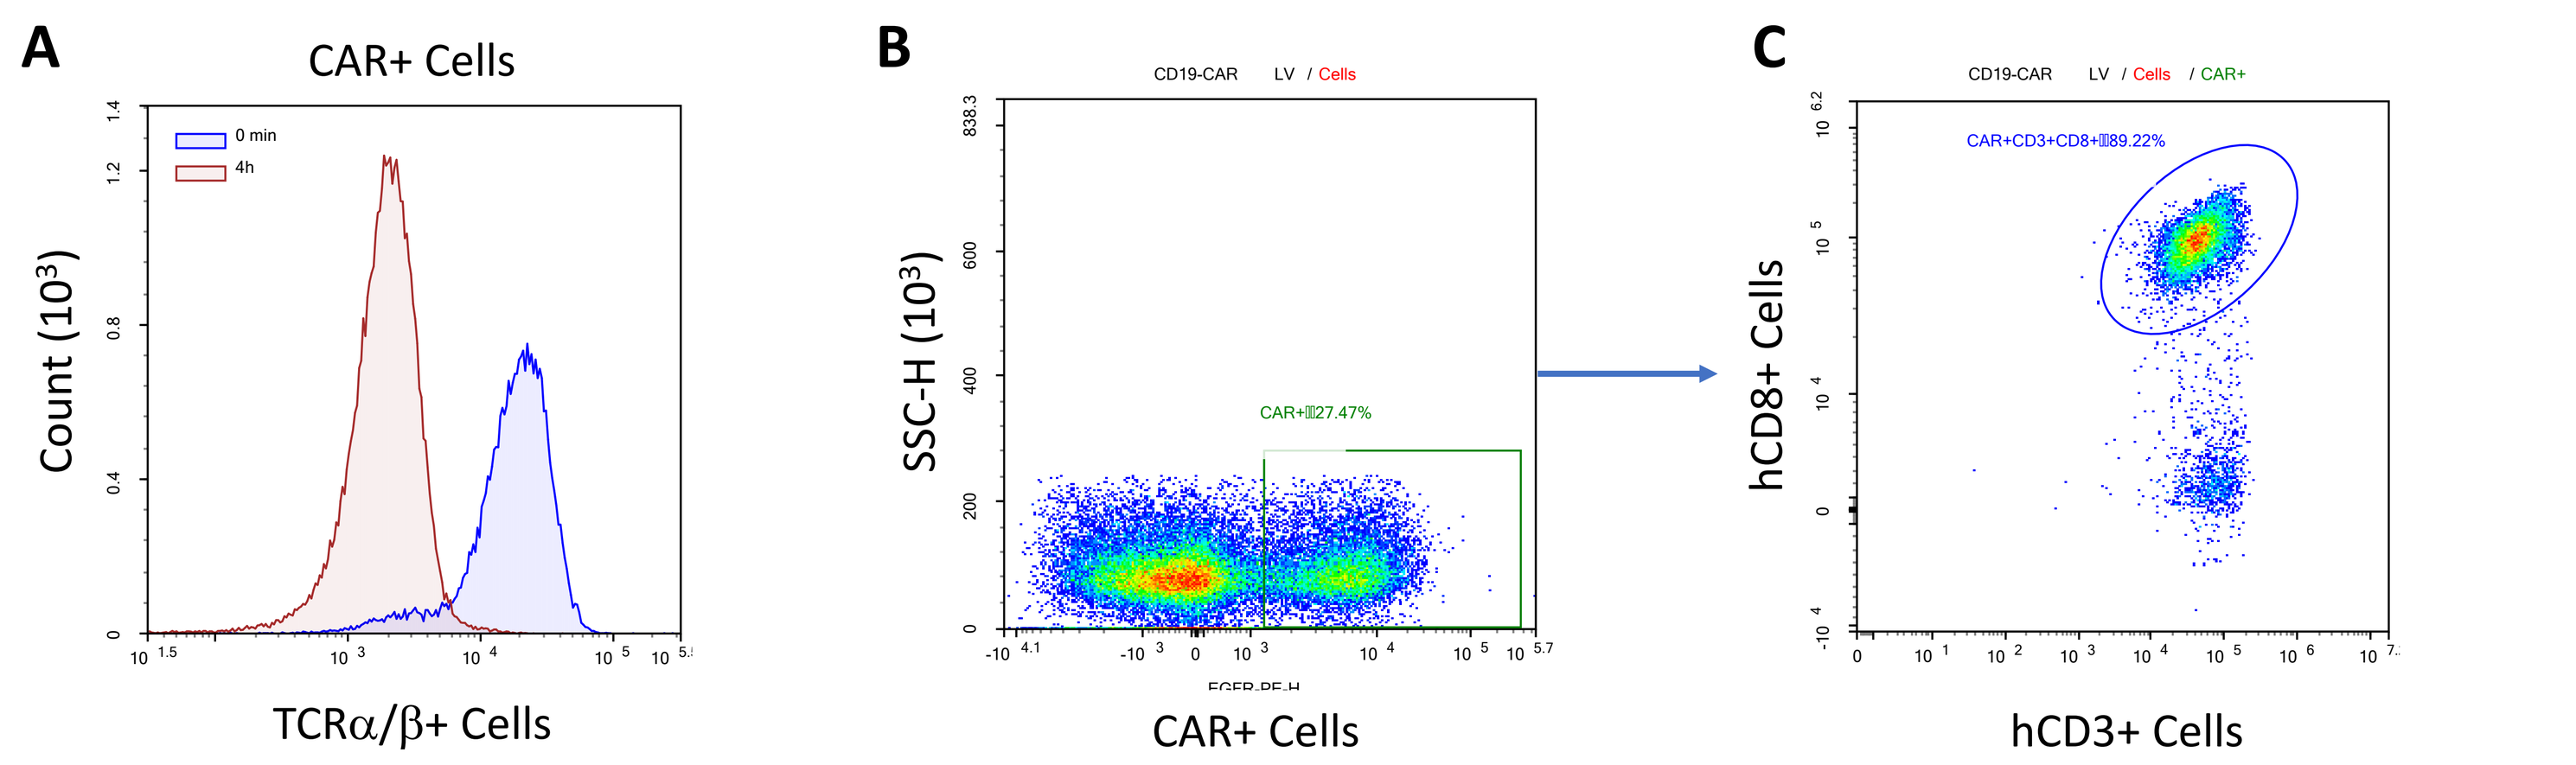
A
CAR+ Cells
0 min
4h
Count (10^3)
TCRα/β+ Cells
B
CD19-CAR LV / Cells
SSC-H (10^3)
CAR+ 27.47%
CAR+ Cells
C
CD19-CAR LV / Cells / CAR+
CAR+CD3+CD8+ 89.22%
hCD8+ Cells
hCD3+ Cells

Fig 1: **A.** Four hours post LV exposure, TCR internalization was observed by flow cytometry, indicating activation of T cells. **B.** LV exposed PBMCs were cultured in GREX plates with IL-2 (10 ng/mL) and IL-7 (10 ng/mL). Twelve days post-transduction, the cells had expanded ~100 times in culture without any additional stimulation and robustly expressed CD19-CAR (>25% CAR+ cells). **C.** The majority (>99%) of CAR+ cells were CD3+, indicating successful transgene integration into T cells. Furthermore, the phenotype of the expanded CD19-CAR+ cells was predominately CD3+CD8+ (>89%).

Results *in vivo*

Figure 2. Humanized Mouse Models

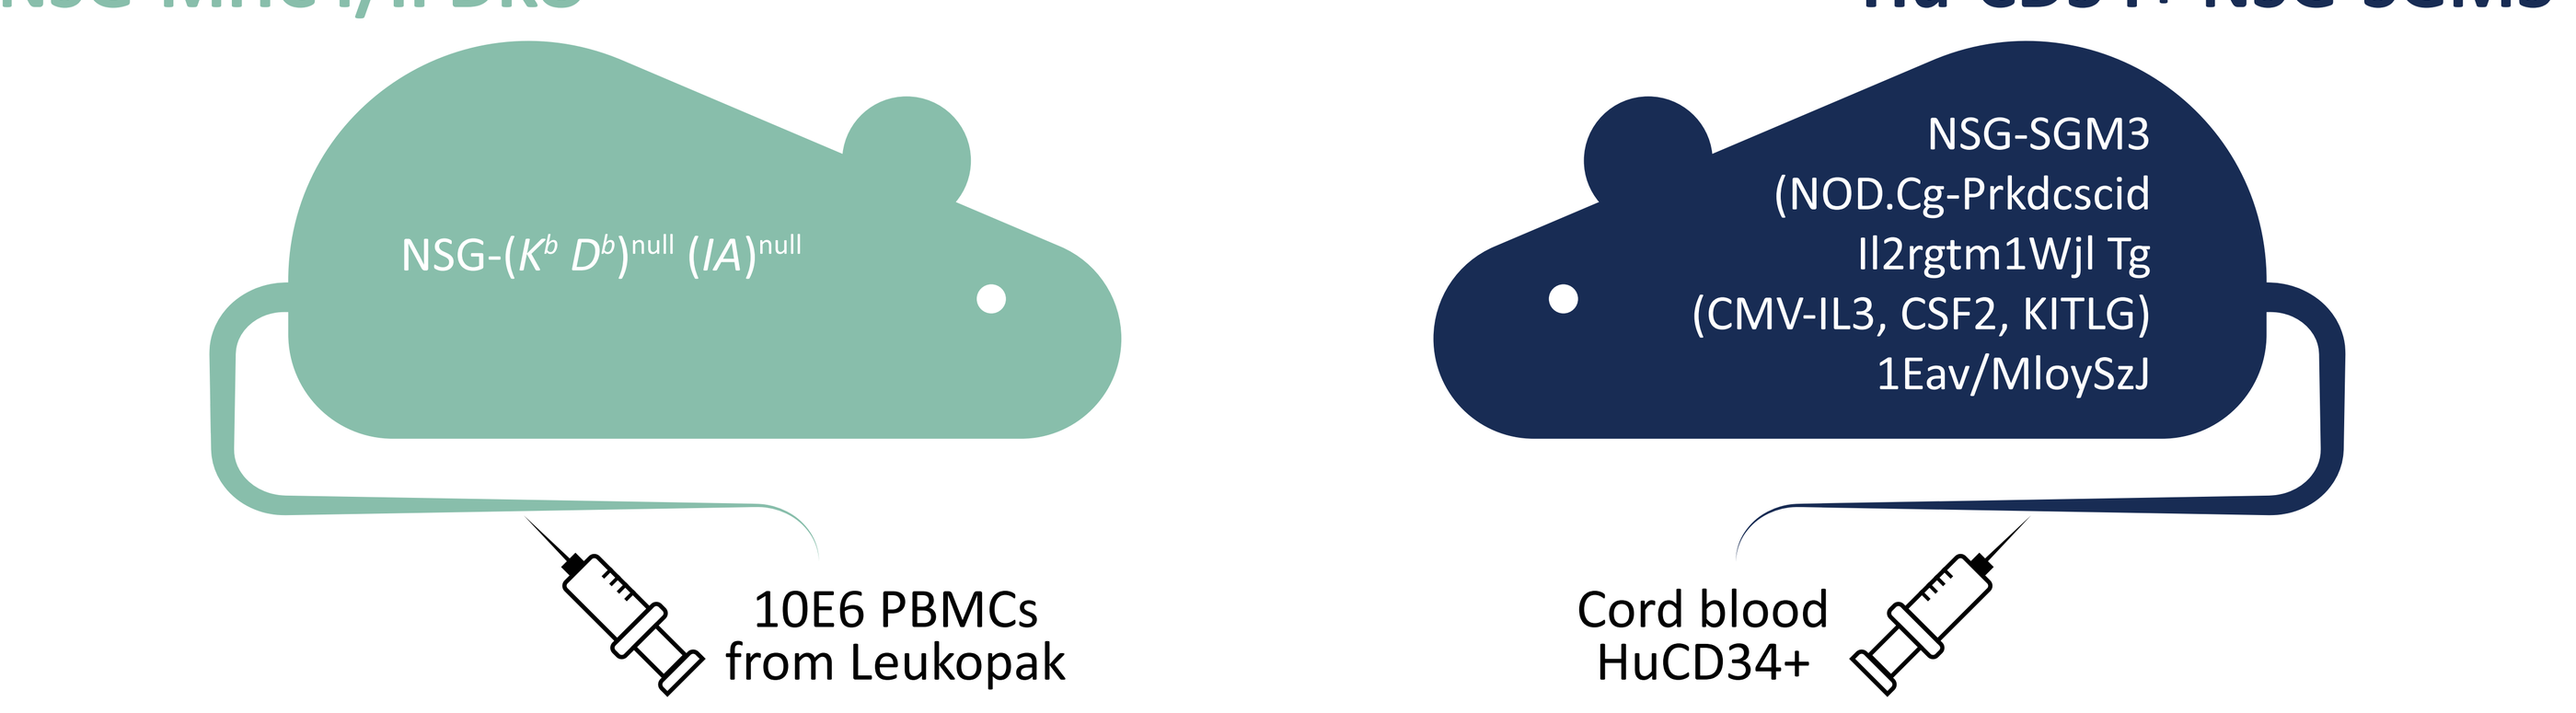
NSG-MHC I/II DKO
NSG-(K^b D^b)^{null} (*IA*)^{null}
10E6 PBMCs from Leukopak
Hu-CD34+ NSG-SGM3
NSG-SGM3 (NOD.Cg-Prkdcscid Il2rgtm1Wjl Tg (CMV-IL3, CSF2, KITLG) 1Eav/MloySzJ
Cord blood HuCD34+

PBMC Composition at Injection

hCD5+	hCD4+	hCD8+	hCD14+
44.95%	25.16%	20.36%	16.52%

hCD15+	hCD20+	hCD56+
25.36%	3.43%	11.58%

SGM3 Blood Composition / Human Chimerism at Day 0

hCD45+	hCD3+	hCD20+	hCD14+	hCD15+
4.89%-34.40%	20.57%-33.00%	16.02%-45.88%	17.67%-49.31%	1.24%-5.83%

Fig 2: Generation of humanized mouse models. MHC Class I and II double knock-out (DKO, JAX stock #025216) mice were injected with 10E6 PBMC isolated from a Leukopak, the cell composition is indicated. SGM3 mice (JAX stock #013062) were humanized at Jackson Laboratories by injection of human cord blood CD34+ hematopoietic stem cells following myeloablation and given 12 weeks for full engraftment prior to the start of this study. The cell composition at D0 of this study is indicated.

Figure 3. Evidence for Generating hCD3+ CAR+ Cells *In Vivo*

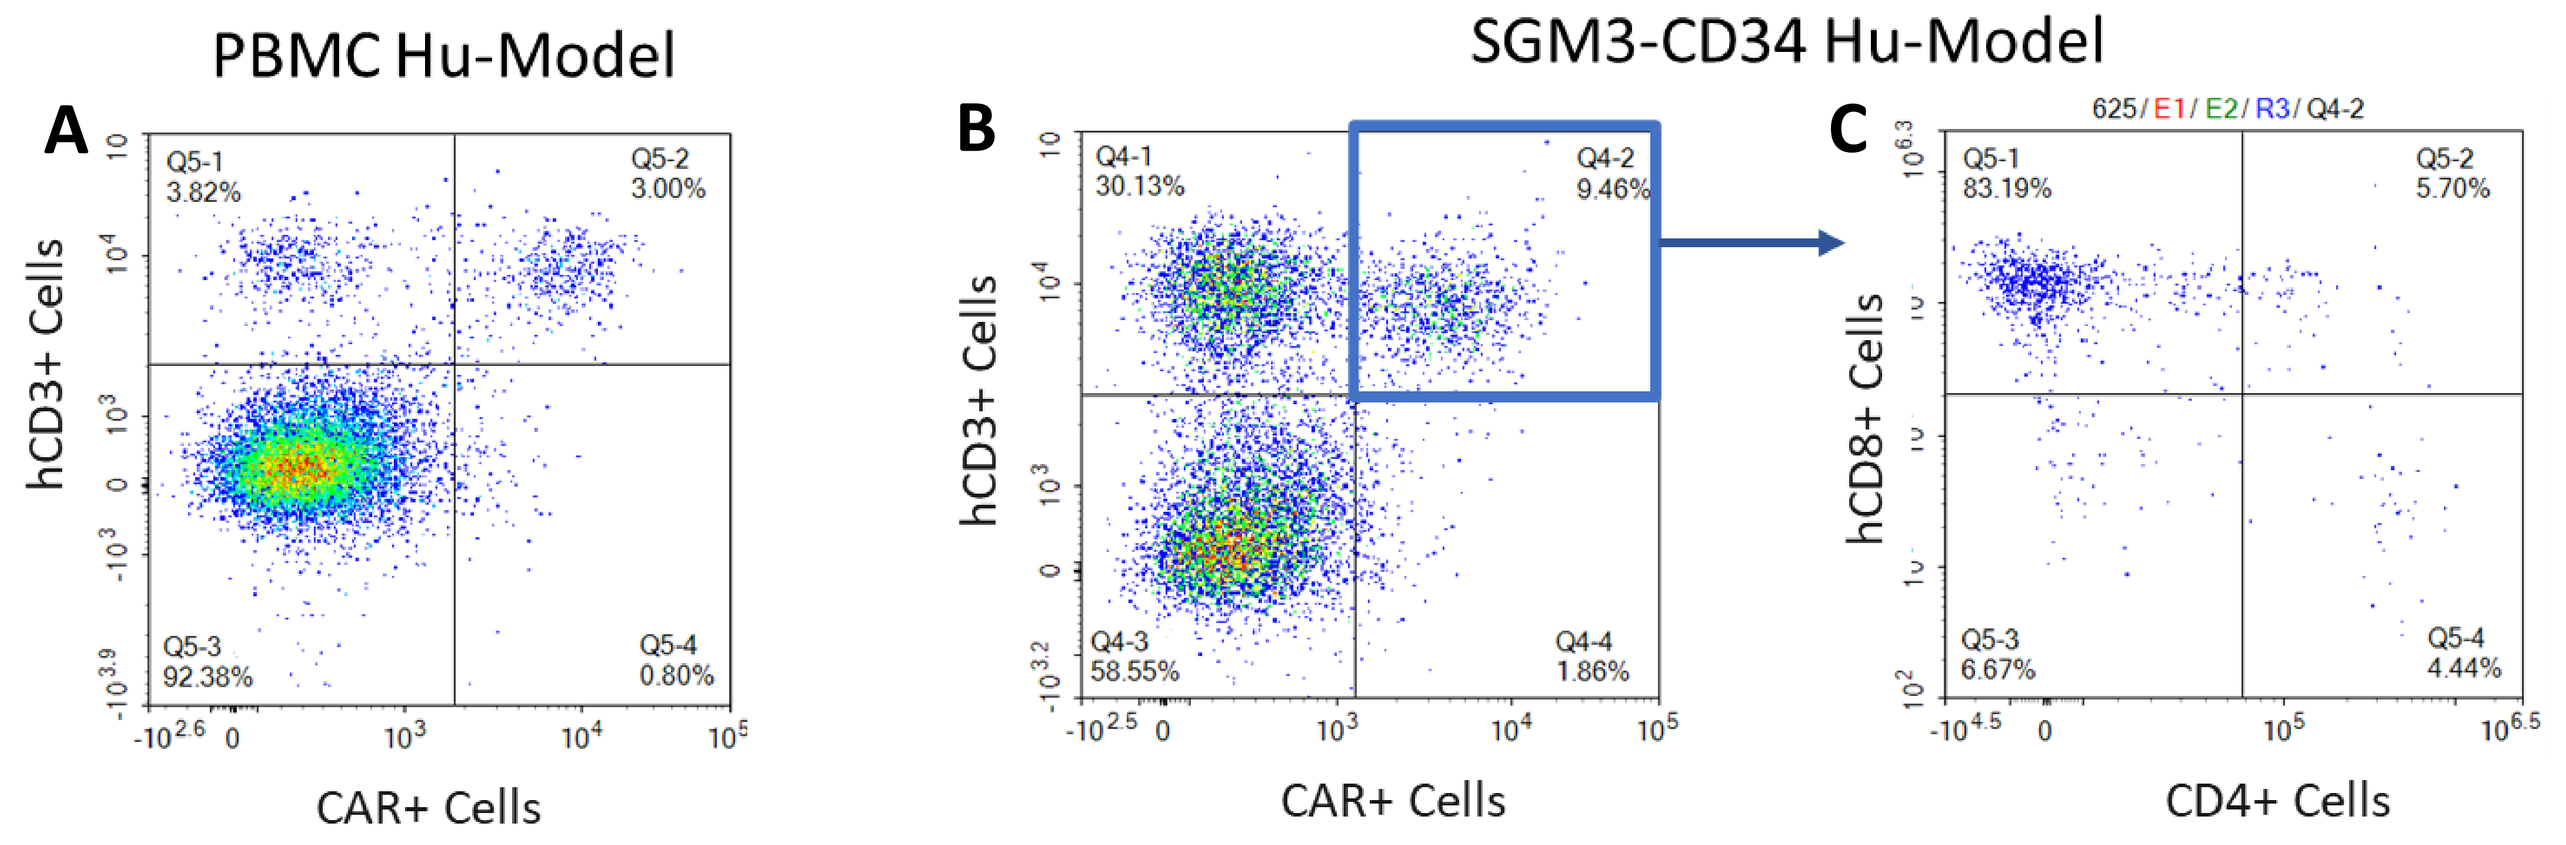
PBMC Hu-Model
SGM3-CD34 Hu-Model
A
Q5-1 3.82%
Q5-2 3.00%
Q5-3 92.38%
Q5-4 0.80%
hCD3+ Cells
CAR+ Cells
B
Q4-1 30.13%
Q4-2 9.46%
Q4-3 58.55%
Q4-4 1.86%
hCD3+ Cells
CAR+ Cells
C
625/ E1/ E2/ R3/ Q4-2
Q5-1 83.19%
Q5-2 5.70%
Q5-3 6.67%
Q5-4 4.44%
hCD8+ Cells
CD4+ Cells

Fig 3: **A**. Flow cytometry reveals the clear presence of hCD3+ CD19-CAR+ cells in peripheral blood in both humanized mouse models 27 days post-injection. **B, C.** Flow cytometry results show that most cells (>80%) within the hCD3+ CAR+ population (**B**) are CD8+CD4- CAR T cells (C)

Figure 4. Evidence for Expansion and Persistence of hCD3+ CAR+ Cells *In Vivo* in Peripheral Blood of Hu-CD34+ NSG-SGM3 Mice

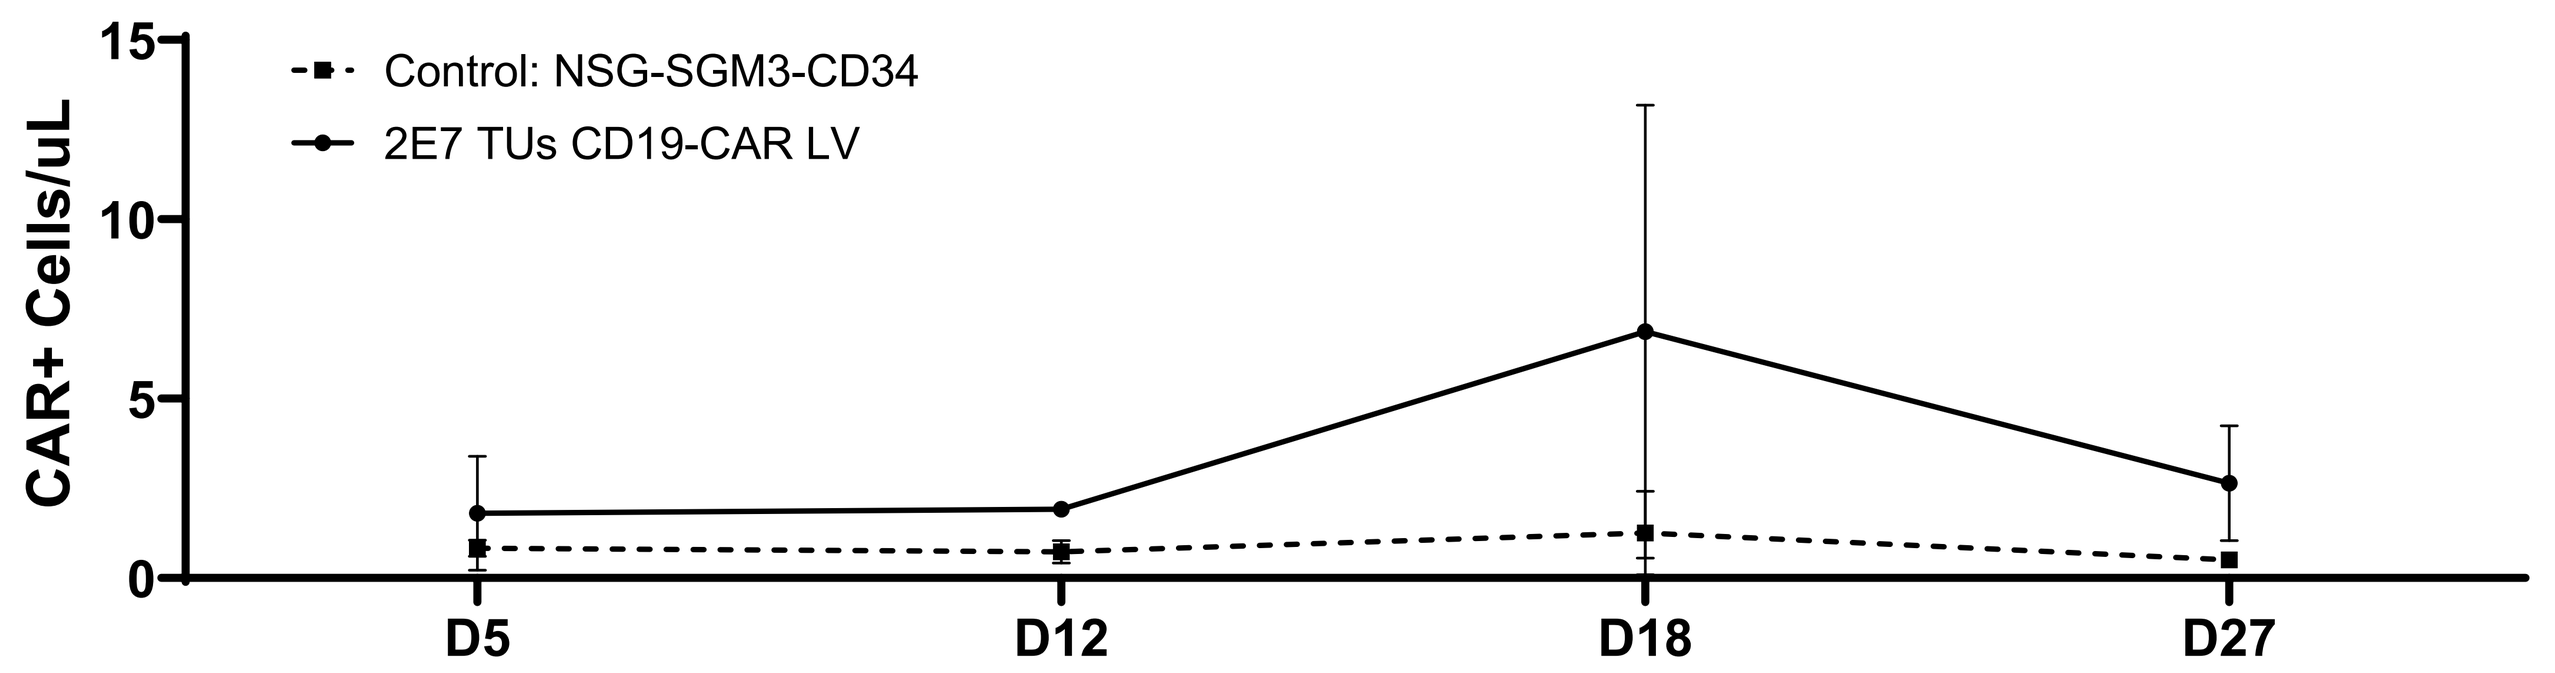
Control: NSG-SGM3-CD34
2E7 TUs CD19-CAR LV
CAR+ Cells/uL
15
10
5
0
D5
D12
D18
D27

Fig 4: Flow cytometry and quantification of hCD45+ hCD3+ CD19-CAR+ cells in peripheral blood of Hu-CD34+ NSG-SGM3 mice at different time points indicate the expansion and persistence of CAR+ cells. The experiment was ended on D27 for histologic evaluation. Mean +/- SD shown, n = 3-6.

Figure 5: Depletion of Circulating Human CD20+ B cells in Peripheral Blood of Hu-CD34+ NSG-SGM3 Mice

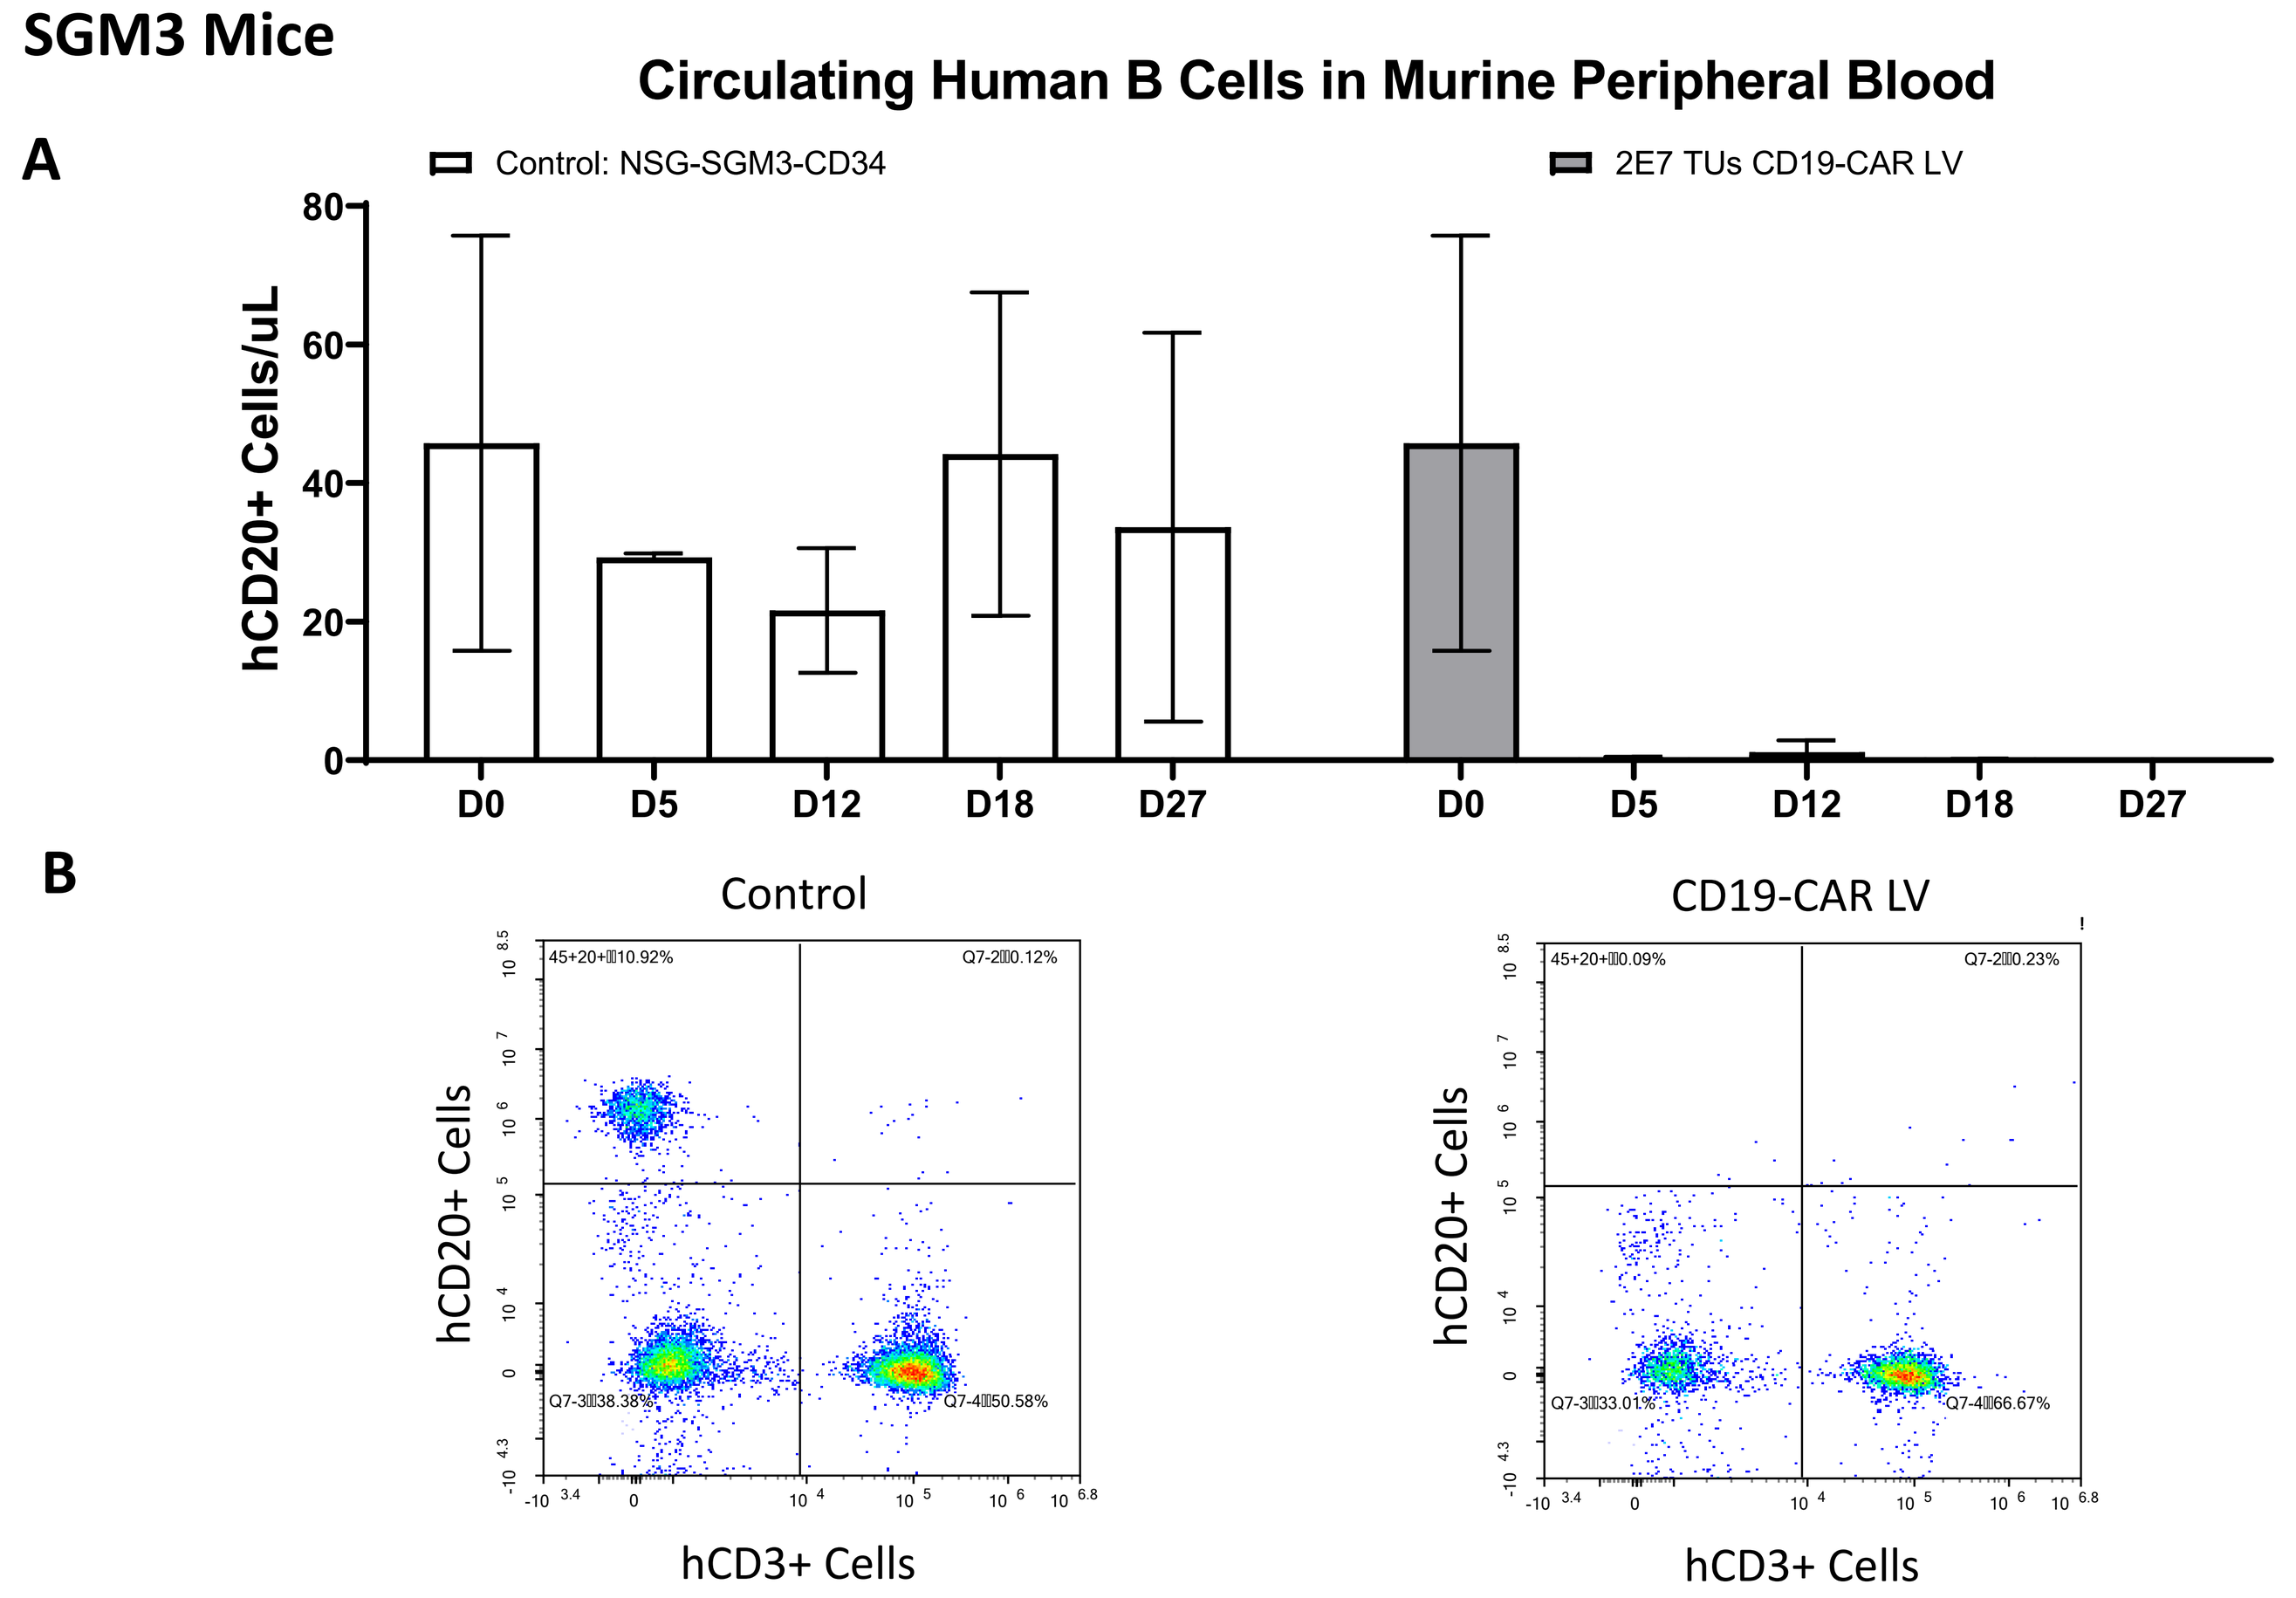
Circulating Human B Cells in Murine Peripheral Blood
A
Control: NSG-SGM3-CD34
2E7 TUs CD19-CAR LV
hCD20+ Cells/uL
80
60
40
20
0
D0
D5
D12
D18
D27
D0
D5
D12
D18
D27
B
Control
CD19-CAR LV
hCD20+ Cells
hCD3+ Cells
hCD20+ Cells
hCD3+ Cells

Fig 5: Flow cytometry results from peripheral blood for hCD20+ cells following direct *in vivo* injection of LV vector encoding CD19-CAR and driver. **A.** hCD20+ cells are not present in the peripheral blood from treated mice (IP 2E7 TUs of CD3-targeted CD19-CAR encoding LV) as early as day 5 vs. persistent hCD20+ cell levels in control mice injected with PBS. This phenotype persisted until the end of the study. Mean +/- SD shown, n = 3-6. **B.** Representative FACS plots for hCD20+ cells at D27.

Figure 6: Depletion of hCD20+ B Cells in MLNs and Spleens of Hu-CD34+ NSG-SGM3 Mice

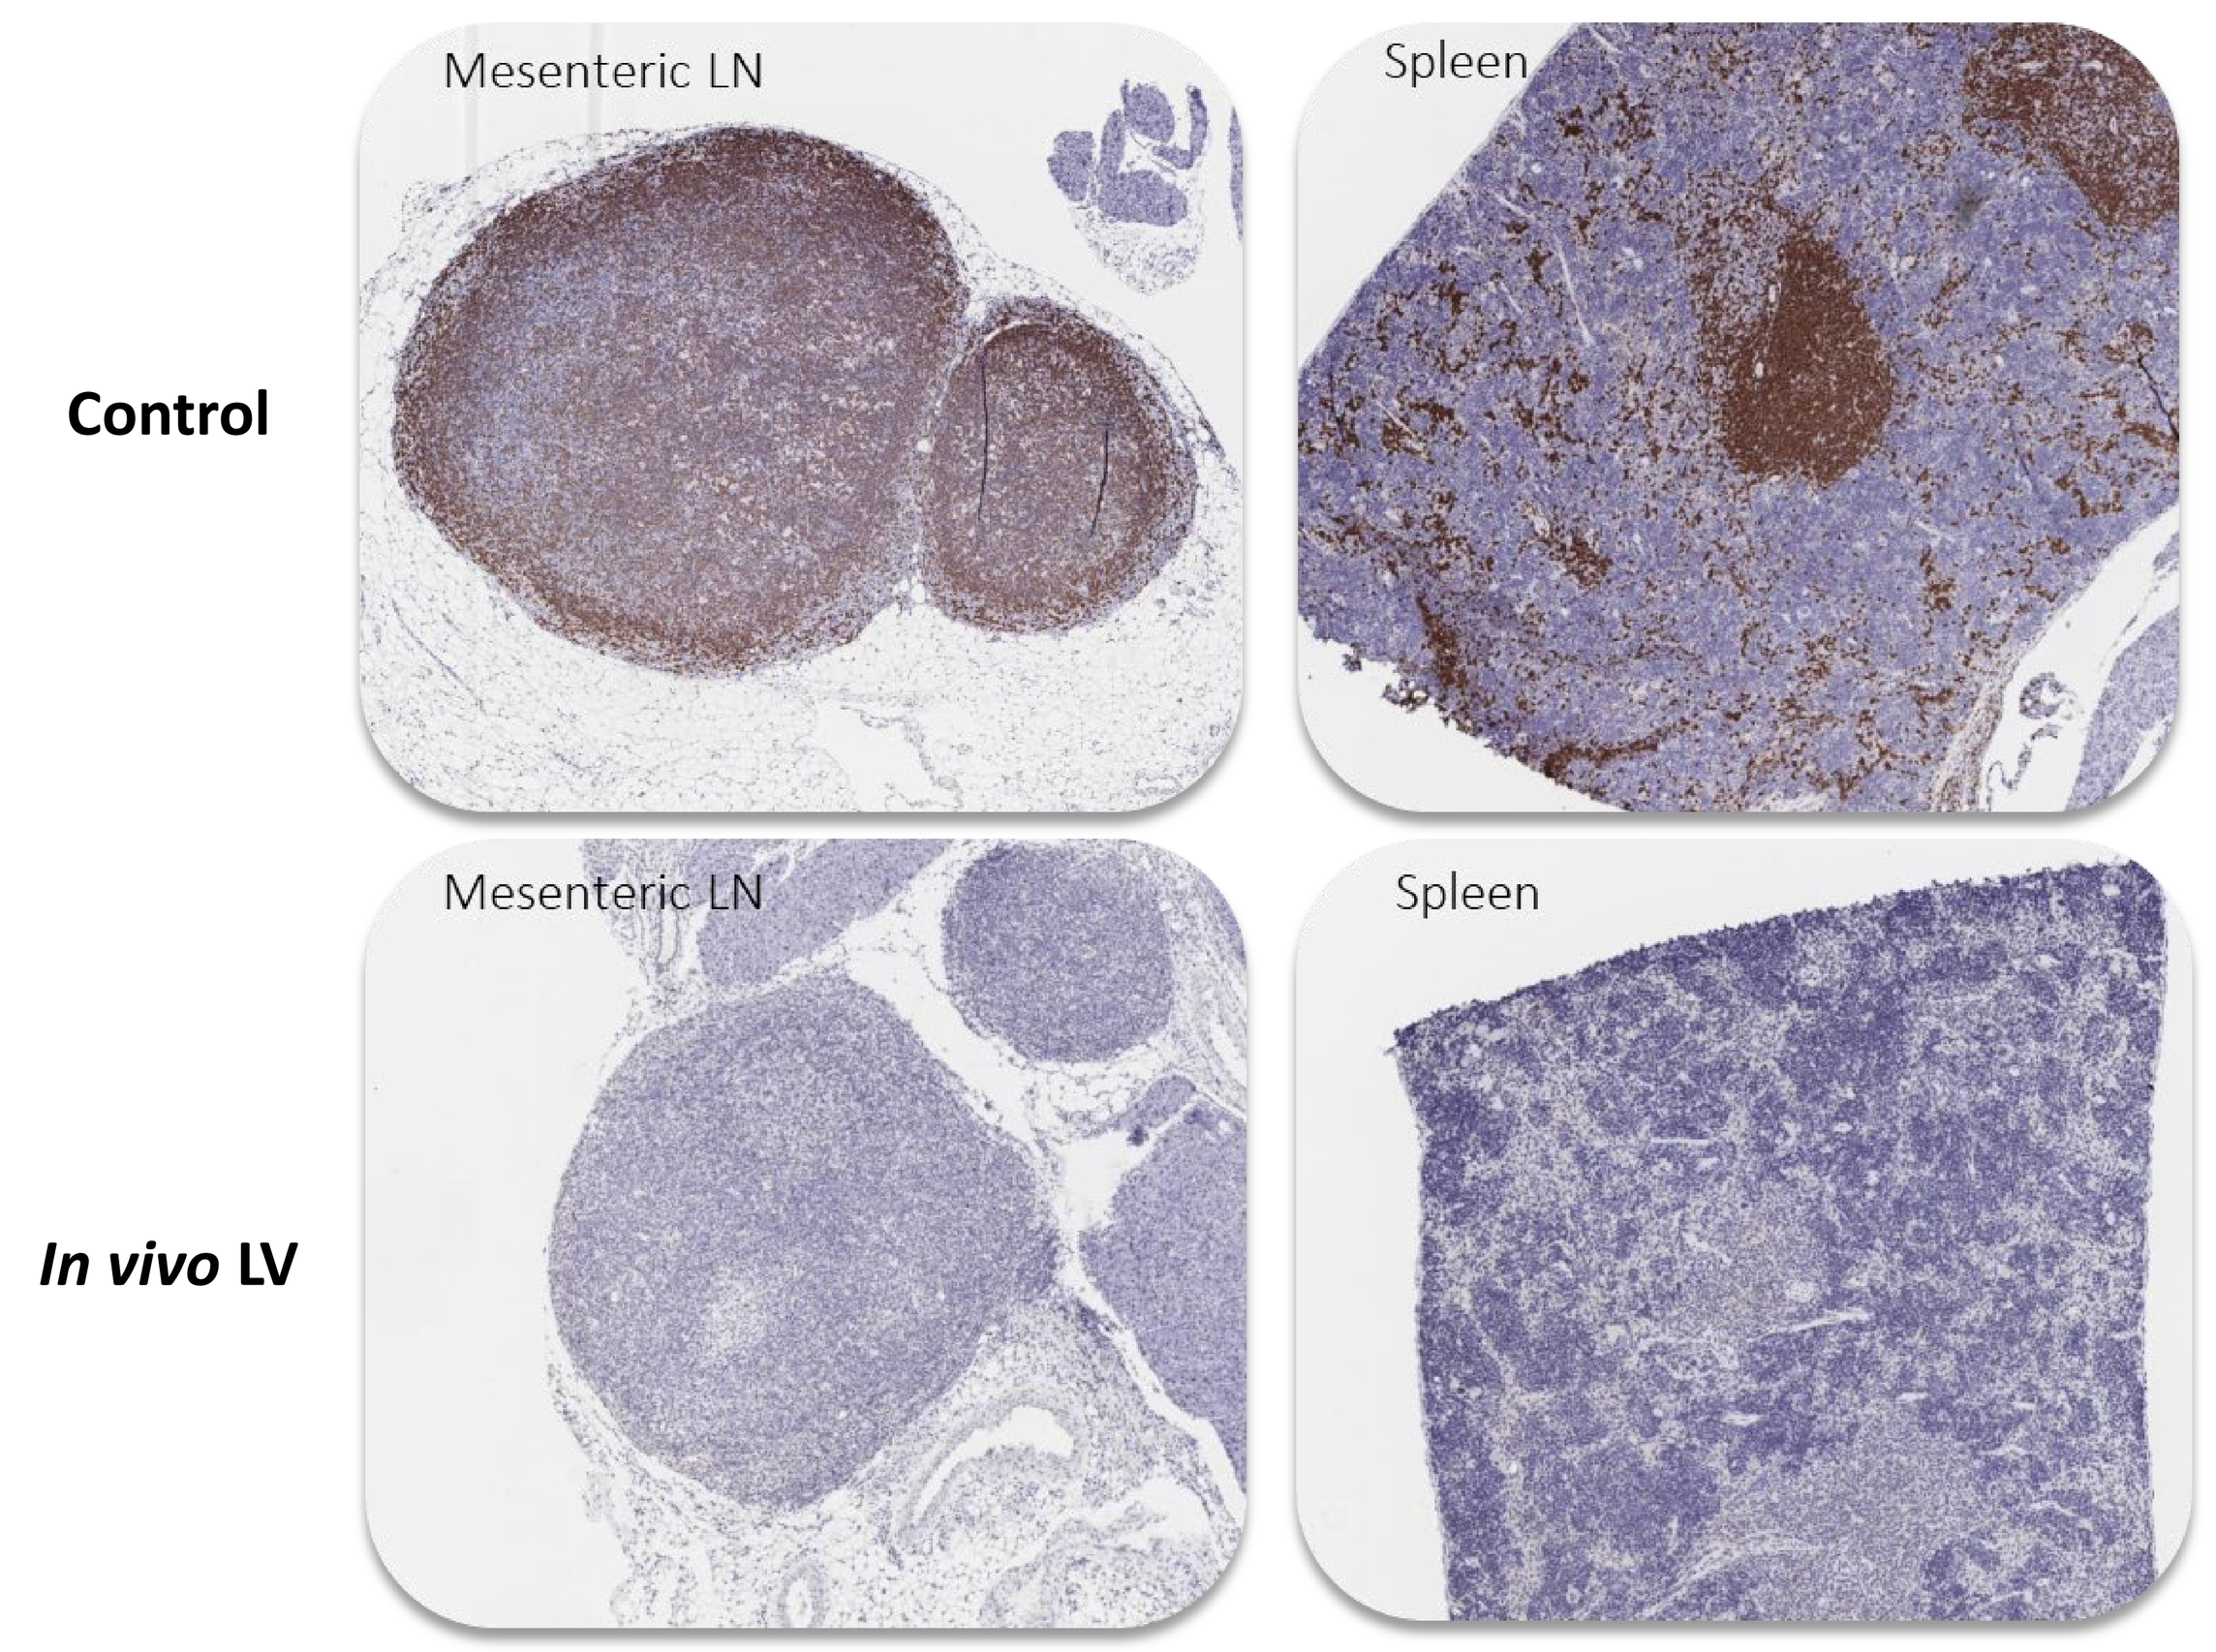
Control
In vivo LV
Mesenteric LN
Spleen
Mesenteric LN
Spleen

Fig 6: Immunohistochemistry images from Mesenteric Lymph Nodes (MLN) and Spleens reveal depleted hCD20+ cells in the treatment groups (IP 2E7 TUs of CD3-targeted CD19-CAR encoding LV) at 27 days post-injection.

Figure 7: qPCR on Mesenteric Lymph Nodes of Hu-CD34+ NSG-SGM3 Mice

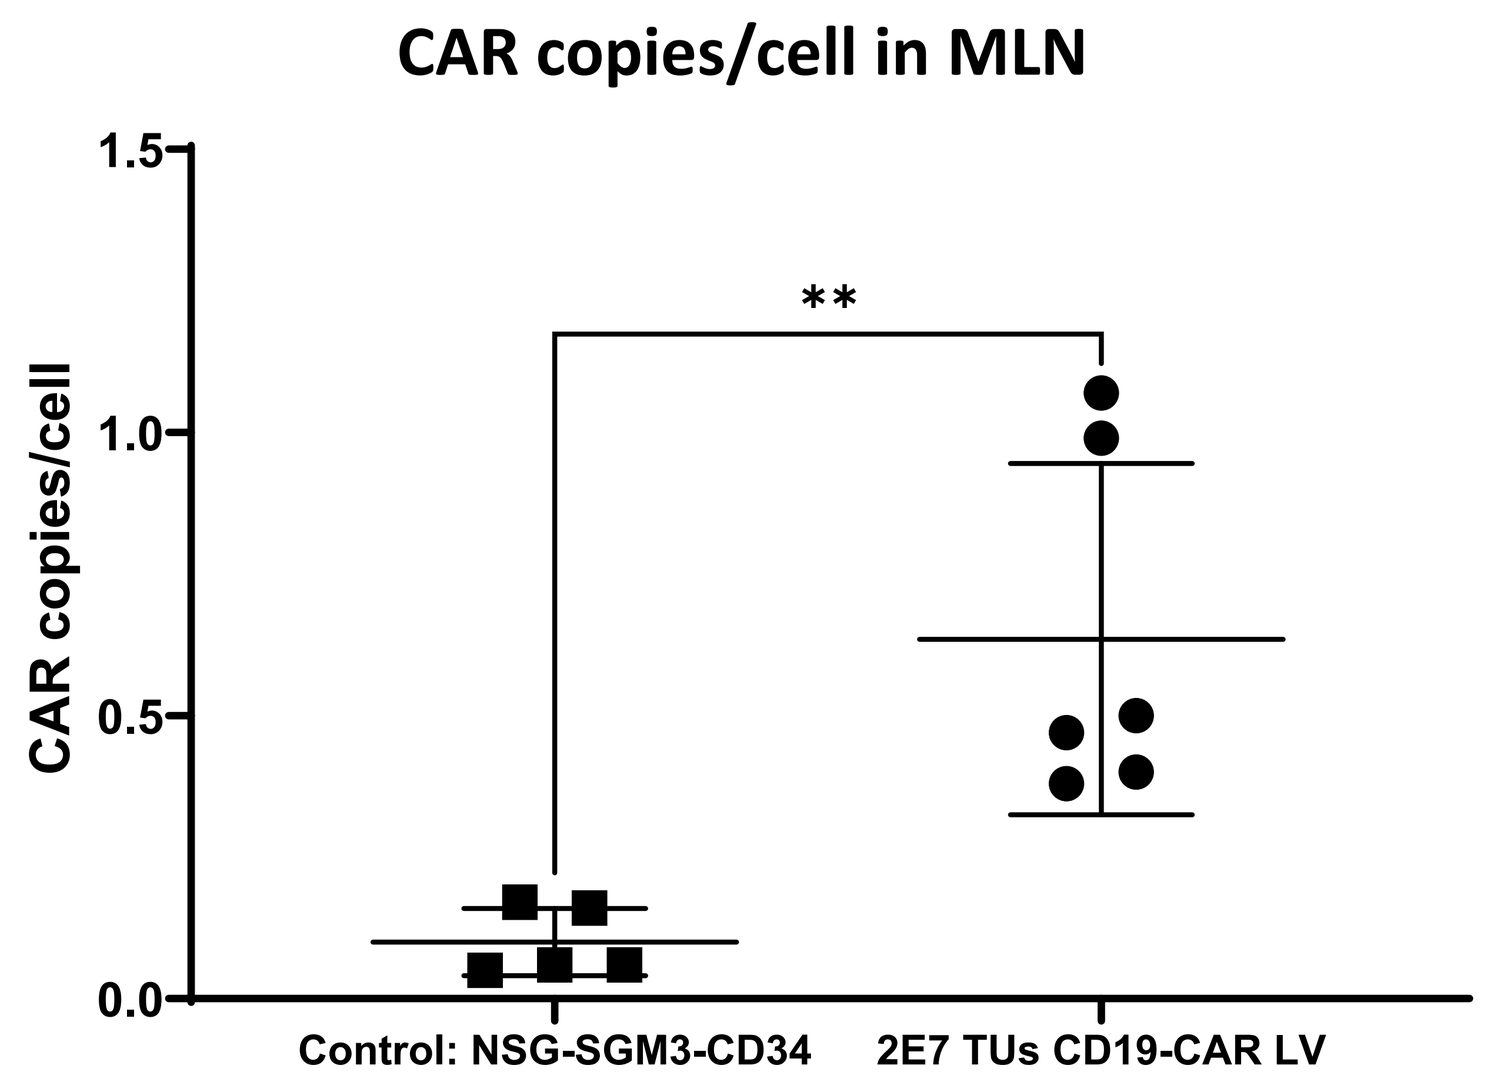
CAR copies/cell in MLN
CAR copies/cell
1.5
1.0
0.5
0.0
**
Control: NSG-SGM3-CD34
2E7 TUs CD19-CAR LV

Fig 7: Quantity of CAR copies per cell using (CAR Copies/ug)/((RNaseP copies/ug)x2). Multiplex qPCR analysis for CAR copies in 50 ng of gDNA extracted from FFPE Mesenteric Lymph Nodes from SGM3 mice 27 days post-injection of CD19-CAR LV. Unpaired T-test was performed, p-value: * < 0.5, ** < 0.01. Mean +/- SD and distribution are shown, n = 5-6.

Conclusions

Here we demonstrate:

- A novel *in vivo* gene therapy approach to generate CAR+ cells without the need for labor-intensive and time-consuming *ex vivo* allogeneic or autologous cell processing
- Rapid B cell aplasia in peripheral blood and in secondary lymphoid organs with a single treatment of LV vector encoding a CD19 CAR and driver in a lymphoreplete animal model
- Evidence of CAR copies within mesenteric lymph nodes, indicating the potential for vector integration into CD3+ T cells at nodal sites
- *In vivo CAR-T* gene therapy holds the potential to significantly reduce the cost and complexity associated with cell therapy manufacturing, accelerate the time to treatment, and expand patient access
- The absence of lymphodepletion may allow for additional clinical strategies including re-treatment, multi-dose treatment, multi-target combinations, or combination with existing standard of care

References

1. Brehm MA., *et al*. FASEB. 2018 Mar; 33(3):3137-3151. doi: 10.1096/fj.201800636R
2. Wunderlich M., *et al*. Leukemia. 2010 Oct;24(10):1785-8. doi: 10.1038/leu.2010.158
3. Billerbeck E., *et al*. Blood. 2011 Mar 17; 117(11):3076-86. doi: 10.1182/blood-2010-08-301507
4. Coughlan AM., *et al*. Stem Cells Dev. 2016 Apr 1;25(7):530-41. doi: 10.1089/scd.2015.0289
5. Bryce PJ., *et al*. J Allergy Clin Immunol. 2016 Sep;138(3):769-779. doi: 10.1016/j.jaci.2016.01.049
6. Zufferey R., *et al*. Nat Biotechnol. 1997 Sep; 15(9):871-5. doi: 10.1038/nbt0997-871

Acknowledgements

We would like to thank Histowiz for histology support, Florida Atlantic University for *in vivo* vivarium support, and Jackson Laboratories for Mice. We also would like to thank Greg Wade, Chief Business Officer at EXUMA Biotech for edits.